Delivering for DMD: Leveraging the FORCE platform to deliver exon skipping oligonucleotides for a broad set of DMD patients

CODY A DESJARDINS, John Hall, Reshmii Venkatesan, Pei-yi Shen, Nick Yoder, Tim Weeden, Sihyung Yang, Babak Basiri, John W. Davis II, Stefano Zanotti, Ranjan Batra, Oxana Beskrovnaya

Dyne Therapeutics Inc., Waltham, MA, USA

View on the Dyne Therapeutics publications website:

BACKGROUND

Duchenne muscular dystrophy (DMD) is a rare, X-linked, progressive neuromuscular disease that is characterized by progressive loss of muscle function due to loss of dystrophin expression. Mutations in the *DMD* gene interrupt the open reading frame of the DMD messenger ribonucleic acid (mRNA), resulting in a premature stop codon leading to destruction of the transcript[1]. The resulting loss of dystrophin expression leads to muscle cell membrane damage upon cell contraction and, ultimately, muscle cell death. Without therapeutic intervention, mortality in patients with DMD is typically observed in the second or third decade of life from respiratory or cardiovascular manifestations of DMD[2,3]. Additionally, central nervous system (CNS) manifestations of the disease are also common, with up to half of patients with DMD experiencing neuropsychiatric symptoms or at least one behavioral comorbidity, including depression, anxiety, attention deficiency and hyperactivity disorder, autism, and intellectual or reading disabilities[4].

Currently approved exon skipping therapies for DMD use phosphorodiamidate morpholino oligomers (PMO) to restore the reading frame of the dystrophin pre-mRNA and enable translation of near-full length, functional dystrophin. PMO therapies are hindered by insufficient distribution to muscles as well as CNS, leading to modest efficacy[5,6]. The FORCE platform was designed to solve the delivery challenges of therapeutic antisense oligonucleotides by leveraging Transferrin Receptor-1 (TfR1) biology with the goal of addressing the underlying cause of rare genetic muscle diseases. Preclinical data in mdx mice and results from the zeleciment rostudirsen clinical program in participants with exon 51 skip amenable DMD demonstrate that the FORCE platform enhances PMO delivery to muscle, thereby leading to functional improvement.

Figure 1. Dyne's exon skipping targets genotypes present in >35% of US DMD Population

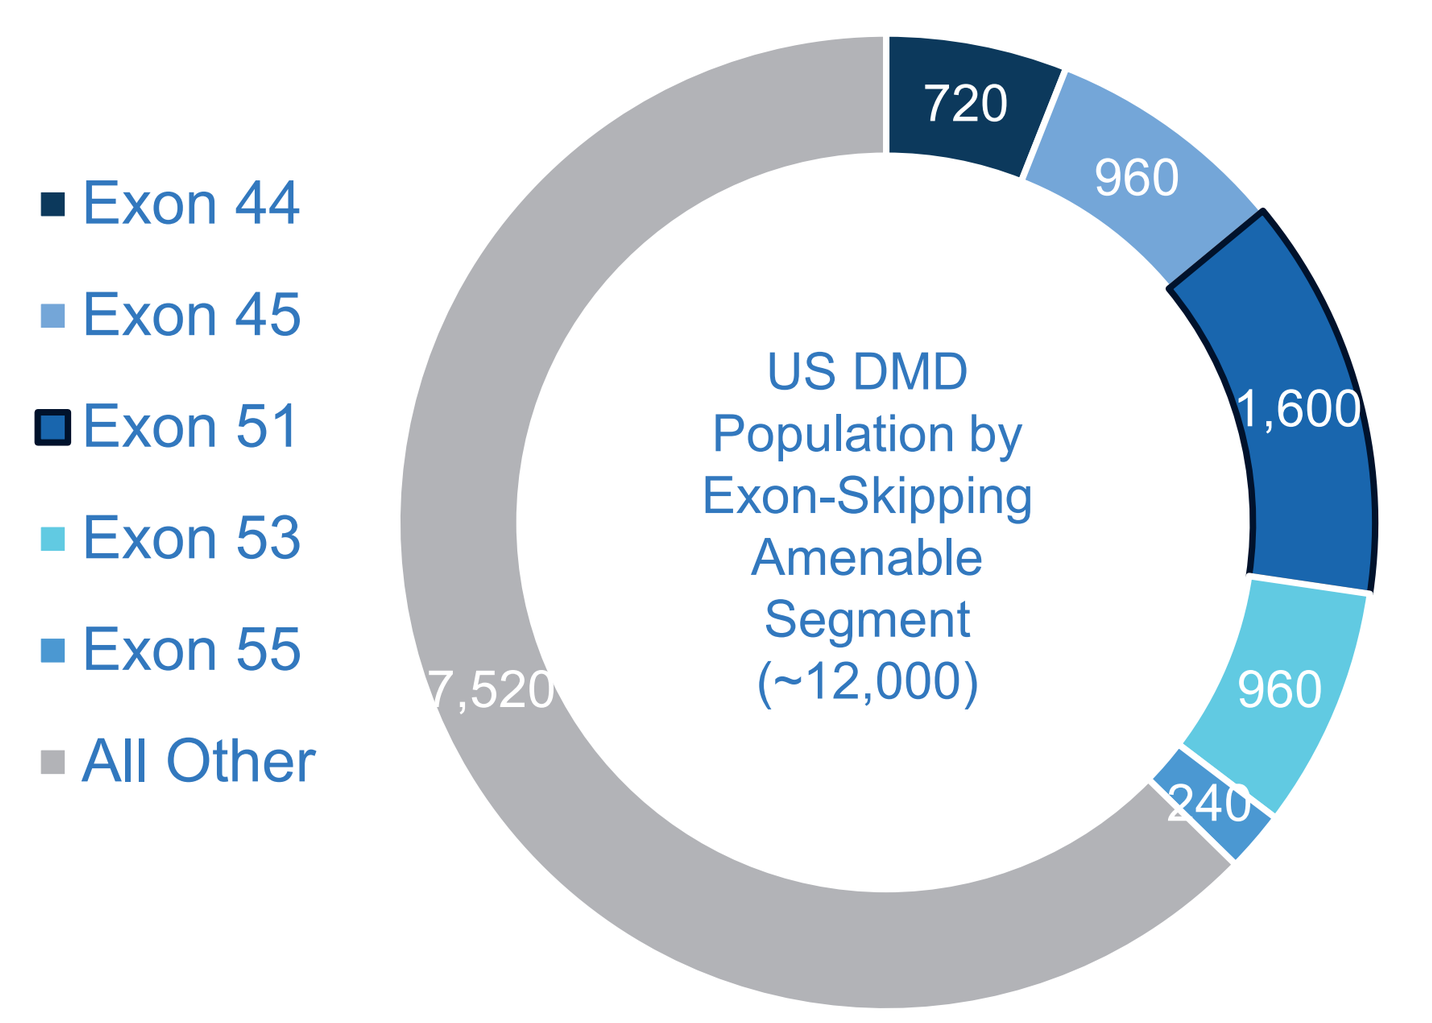
Exon 44
Exon 45
Exon 51
Exon 53
Exon 55
All Other
US DMD Population by Exon-Skipping Amenable Segment (~12,000)
720
960
1,600
960
240
7,520

	Exon 51	Exon 53	Exon 45	Exon 44	Exon 55
% of DMD	13%	8%	8%	6%	2%
US Diagnosed Prevalence	~1,600	~1,000	~1,000	~750	~240
FORCE Conjugate	Z-rostudirsen	DYNE-253	DYNE-245	DYNE-244	DYNE-255

METHODS

Dyne Therapeutics' exon skipping conjugates were designed with our proprietary FORCE™ platform, which consists of 3 key components: 1) a Fab specific for TfR1, engineered to improve the delivery of a therapeutic molecule to skeletal, cardiac, and smooth muscle, as well as the CNS; 2) a linker to conjugate the Fab to the payload; and 3) a therapeutic oligonucleotide as a payload[7]. The payload in Dyne's exon skipping conjugates are antisense PMOs designed to induce skipping of exons 53, 45, 44, or 55, respectively. When the PMO is delivered to cells expressing a mutant *DMD* transcript amenable to exon skipping, the *DMD* reading frame is restored, thereby enabling translation of an internally shortened but functional dystrophin protein[8]. Conjugation of the PMO payload via a valine-citrulline (Val-Cit) linker to the TfR1-targeting Fab enhances delivery to muscle and brain[9, 10]. Dyne's exon skipping conjugates are designed to be internalized into the endosome upon binding to TfR1 and to subsequently release the PMO payload into the cytosol after cleavage of the Val-Cit linker during endosomal trafficking[11].

Payloads designed to skip exons 44, 45, 53, or 55 of the *DMD* transcript were conjugated using the FORCE platform and skipping potential was evaluated in disease-relevant cell culture models of DMD. Subsequently, delivery and pharmacodynamic effects of DYNE-253 were further investigated in the humanized hTfR1 x $hDMD_{WT}$/*mdx* and in the DMD disease relevant hTfR1 x $hDMD_{Del52}$/*mdx* mouse model amenable for exon 53 skipping.

Figure 2. Dyne FORCE Platform Leverages TfR1 to Deliver Exon Skipping Payloads

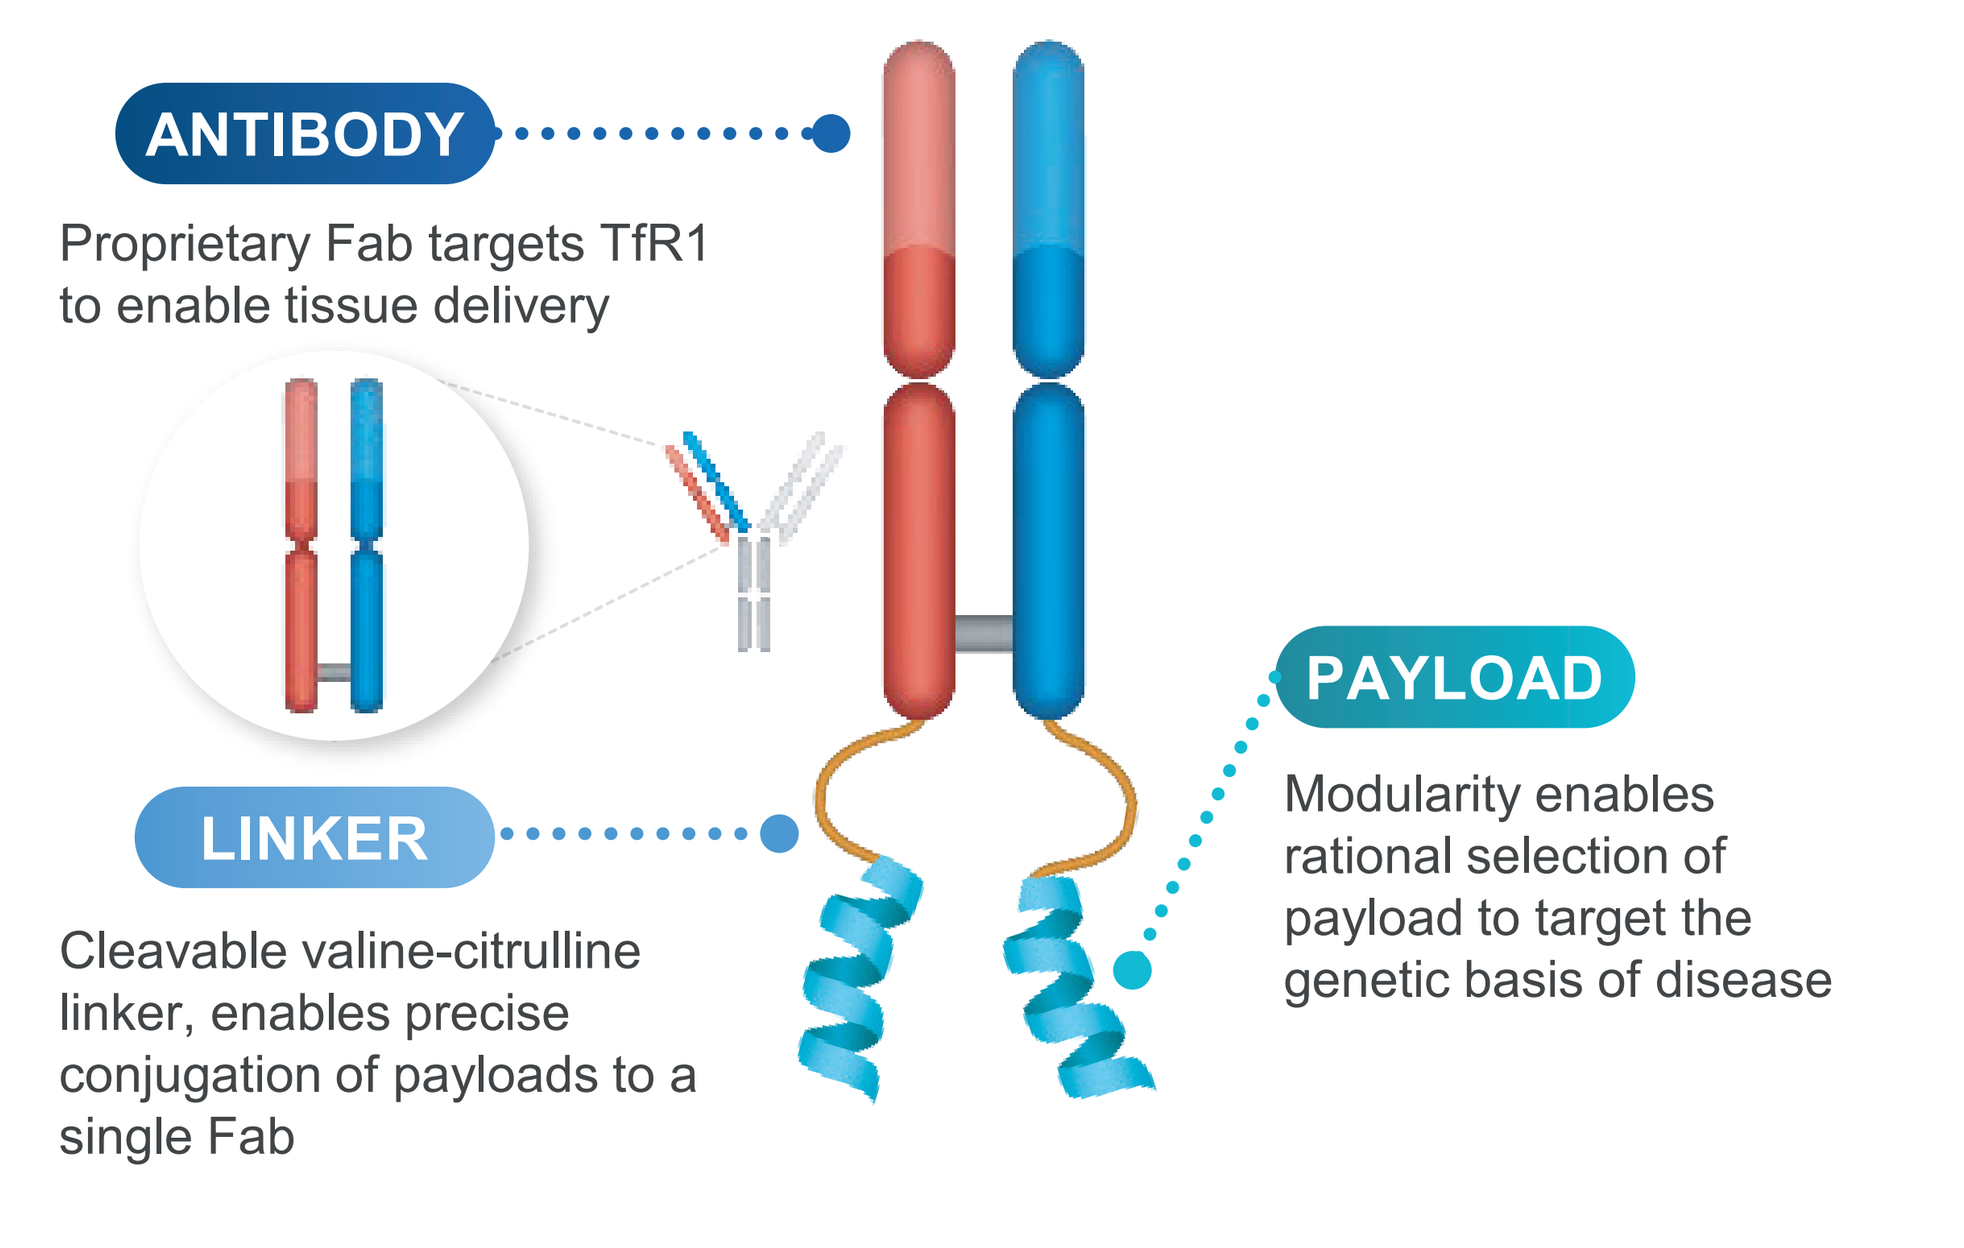
ANTIBODY
Proprietary Fab targets TfR1 to enable tissue delivery
LINKER
Cleavable valine-citrulline linker, enables precise conjugation of payloads to a single Fab
PAYLOAD
Modularity enables rational selection of payload to target the genetic basis of disease

Mechanism of FORCE Delivery of Exon Skipping PMO

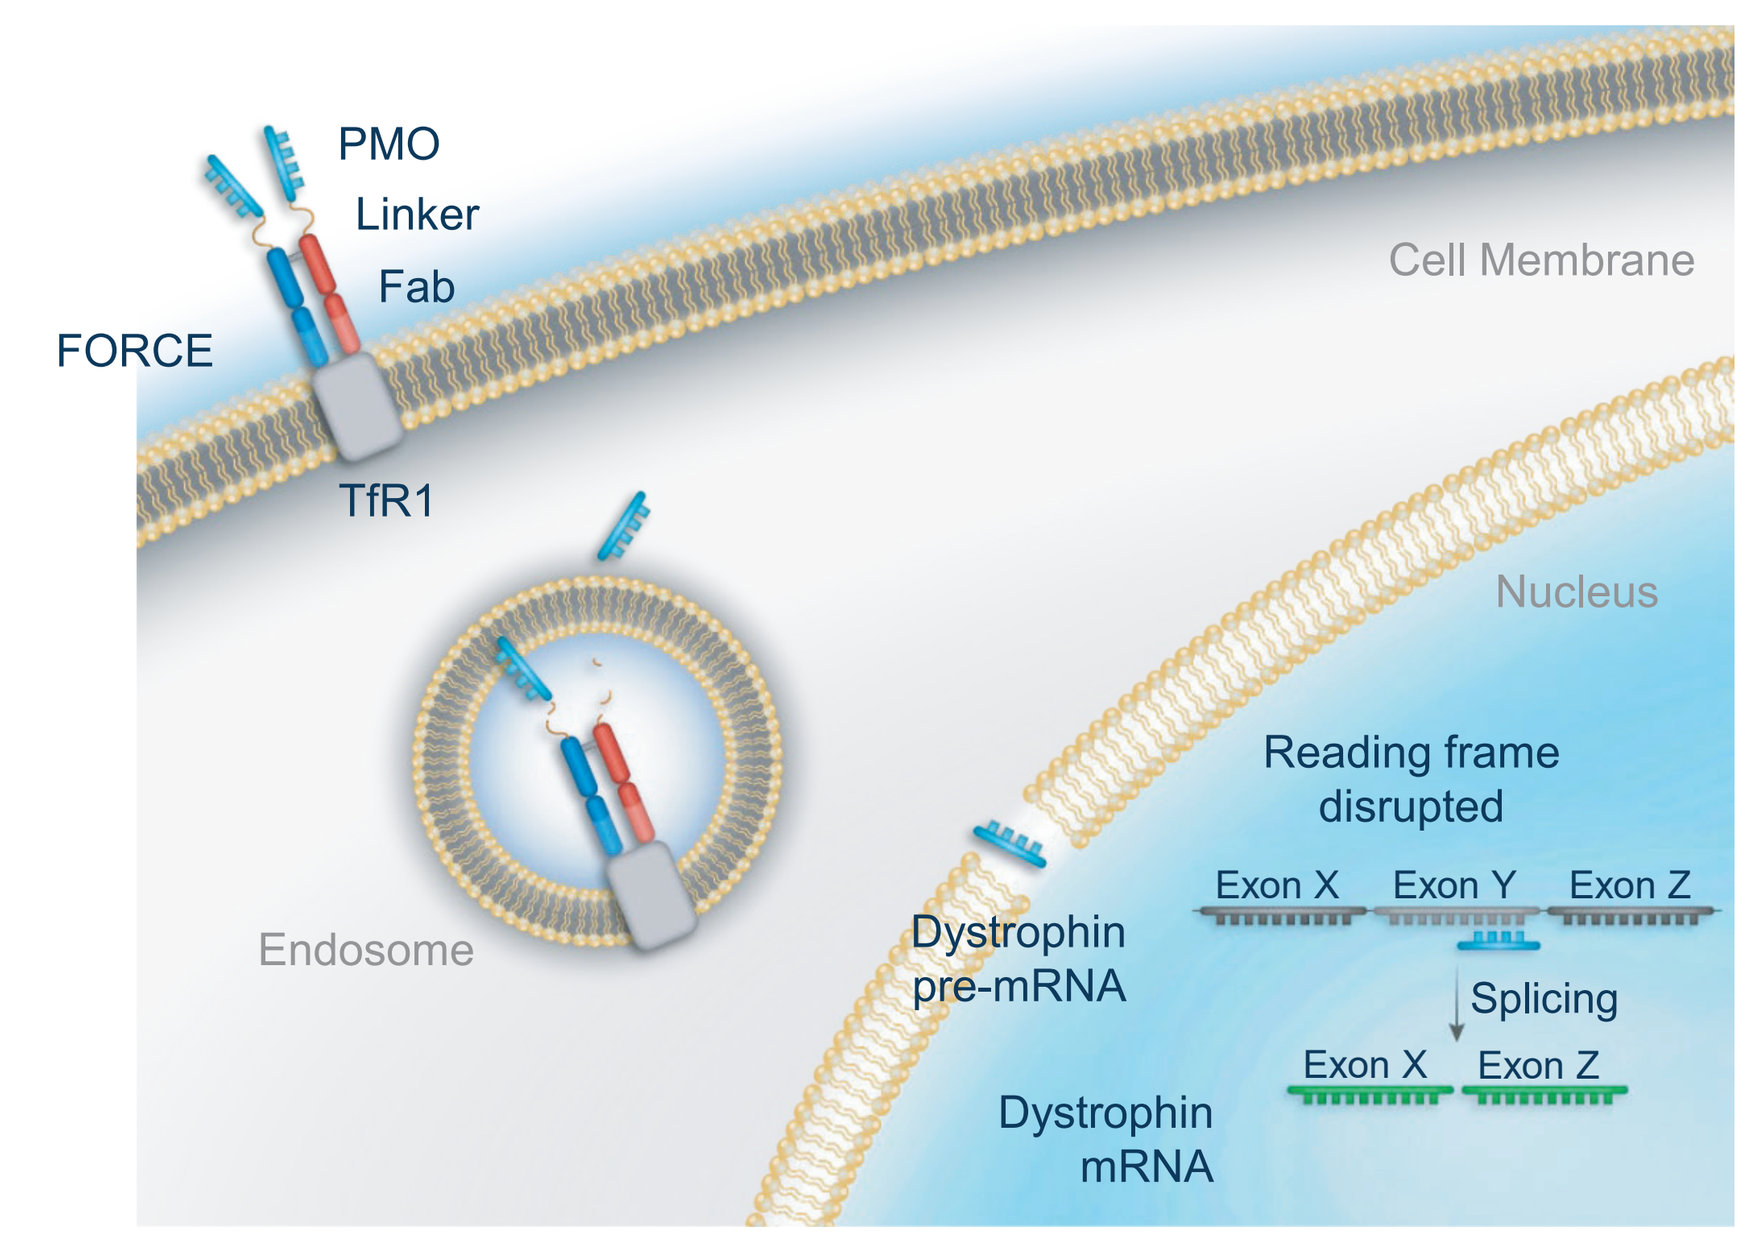
PMO
Linker
Fab
FORCE
TfR1
Cell Membrane
Nucleus
Endosome
Reading frame disrupted
Exon X
Exon Y
Exon Z
Splicing
Dystrophin pre-mRNA
Dystrophin mRNA

RESULTS

Figure 3. FORCE-PMO Conjugates Induce Robust Exon Skipping in DMD Myotubes

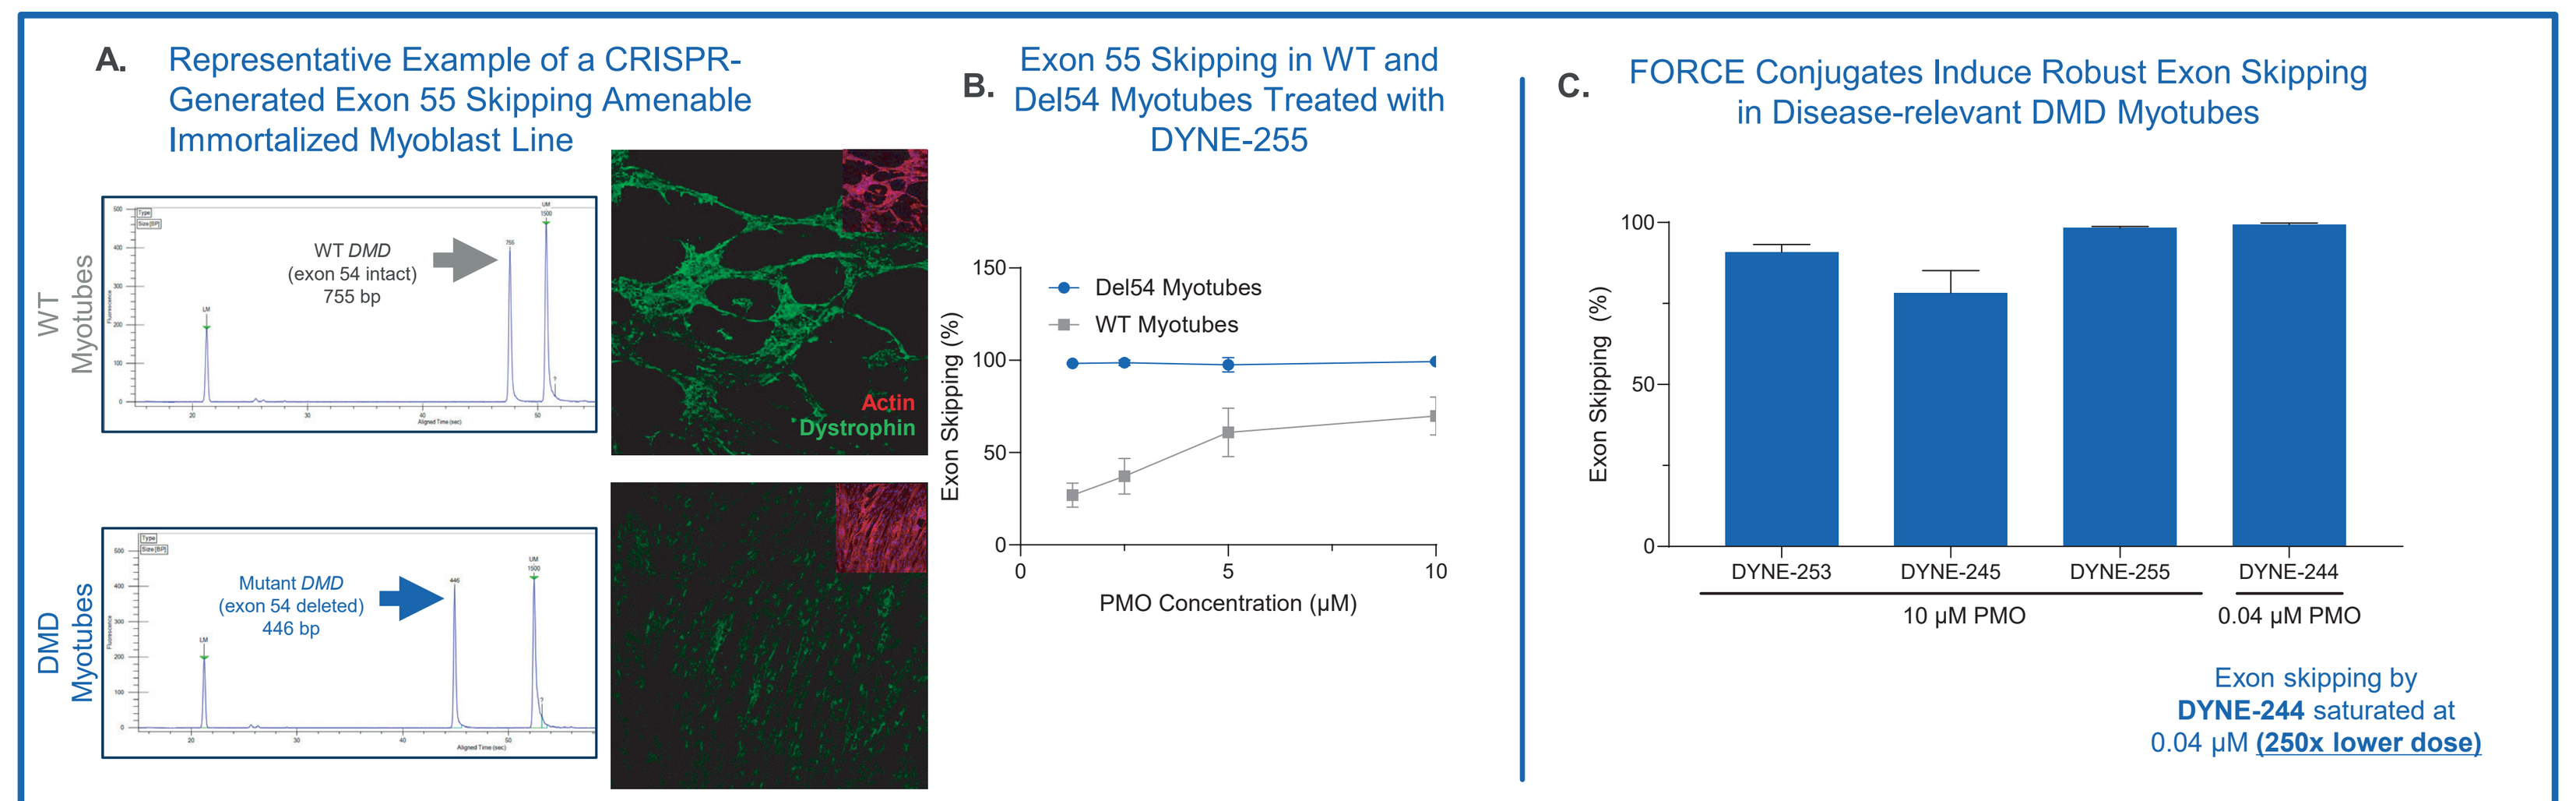
A. Representative Example of a CRISPR-Generated Exon 55 Skipping Amenable Immortalized Myoblast Line
WT Myotubes
WT DMD (exon 54 intact) 755 bp
DMD Myotubes
Mutant DMD (exon 54 deleted) 446 bp
Actin
Dystrophin
B. Exon 55 Skipping in WT and Del54 Myotubes Treated with DYNE-255
Del54 Myotubes
WT Myotubes
Exon Skipping (%)
PMO Concentration (μM)
C. FORCE Conjugates Induce Robust Exon Skipping in Disease-relevant DMD Myotubes
Exon Skipping (%)
DYNE-253
DYNE-245
DYNE-255
DYNE-244
10 μM PMO
0.04 μM PMO
Exon skipping by DYNE-244 saturated at 0.04 μM (250x lower dose)

In vitro assessment of exon skipping confirms the ability of DYNE-253, -245, -255, and -244 to induce exon skipping in cell lines with mutations to skipping of their respective exons. (A) Representative capillary electrophoresis (left panels) and confirmatory dystrophin immunofluorescence (right panels) data supporting loss of dystrophin expression after the deletion of exon 54 of the *DMD* gene in an immortalized myoblast line to generate a myoblast line amenable to *DMD* exon 55 skipping. (B) Exon 55 skipping was confirmed in a dose response study with increasing concentrations of DYNE-255. Notably, exon 55 skipping was substantially more robust in the Del54 myotubes than in the parental WT myotubes. Data are represented as mean ± SD, n=4 wells/group. (C) The ability of DYNE-253, DYNE-245, DYNE-255 (at a 10 μM payload dose), and DYNE-244 (at 0.04 μM were dose) to induce exon skipping was assessed in cell lines bearing mutations amenable to skipping of their respective payloads. Data are represented as mean ± SD, n=3-6 wells/group.

Figure 4. DYNE-253 Enhances PMO Exposure to Tissues of Interest

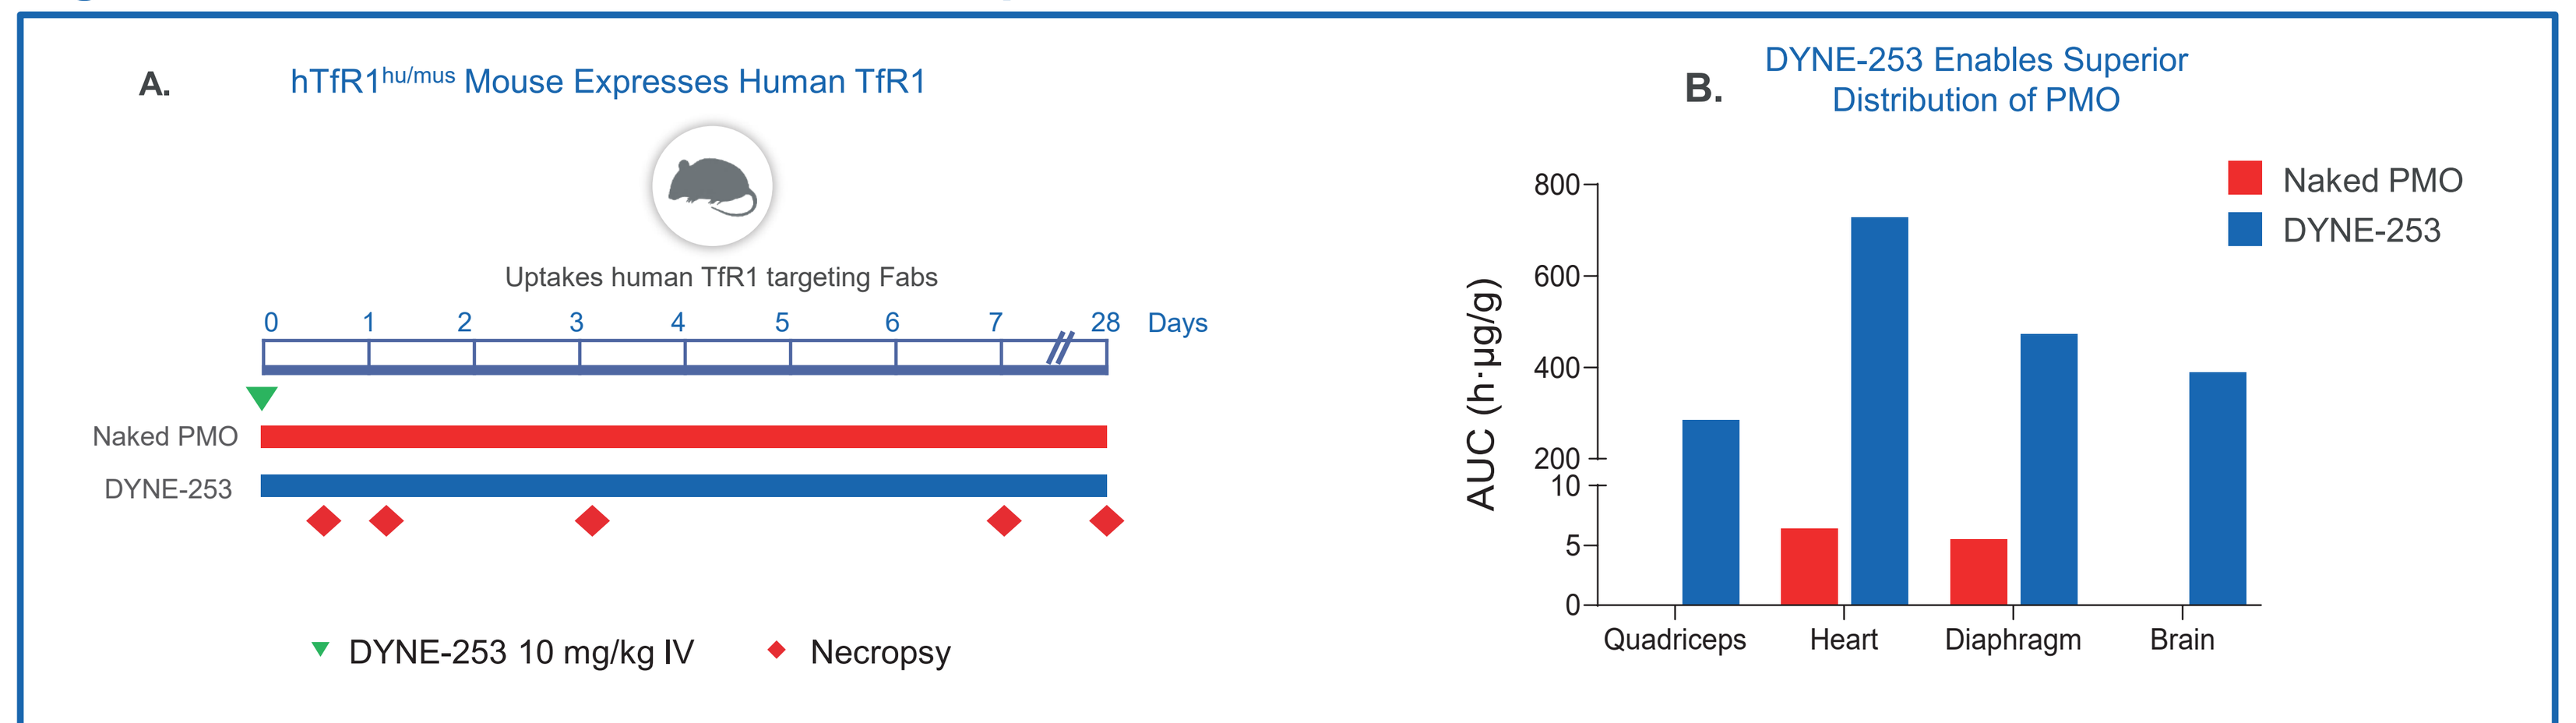
A. hTfR1hu/mus Mouse Expresses Human TfR1
Uptakes human TfR1 targeting Fabs
Days
Naked PMO
DYNE-253
DYNE-253 10 mg/kg IV
Necropsy
B. DYNE-253 Enables Superior Distribution of PMO
Naked PMO
DYNE-253
AUC (h·μg/g)
Quadriceps
Heart
Diaphragm
Brain

DYNE-253 delivers substantially more exon skipping PMO payload to tissues relevant to DMD pathology. (A) Male hTfR1 mice heterozygous for a knock-in of the human TfR1 transcript were administered a single 10 mg/kg IV dose of either DYNE-253 or its PMO payload alone. Animals were necropsied 6h and 1d, 3d, 7d, and 28d post-dose. (B) AUC calculations from tissue exposure data generated at each necropsy support substantially improved delivery to tissues of interest with DYNE-253 compared to the naked PMO payload alone.

Figure 5. Disease-Relevant Del52 Mice are More Responsive to Exon 53 Skipping

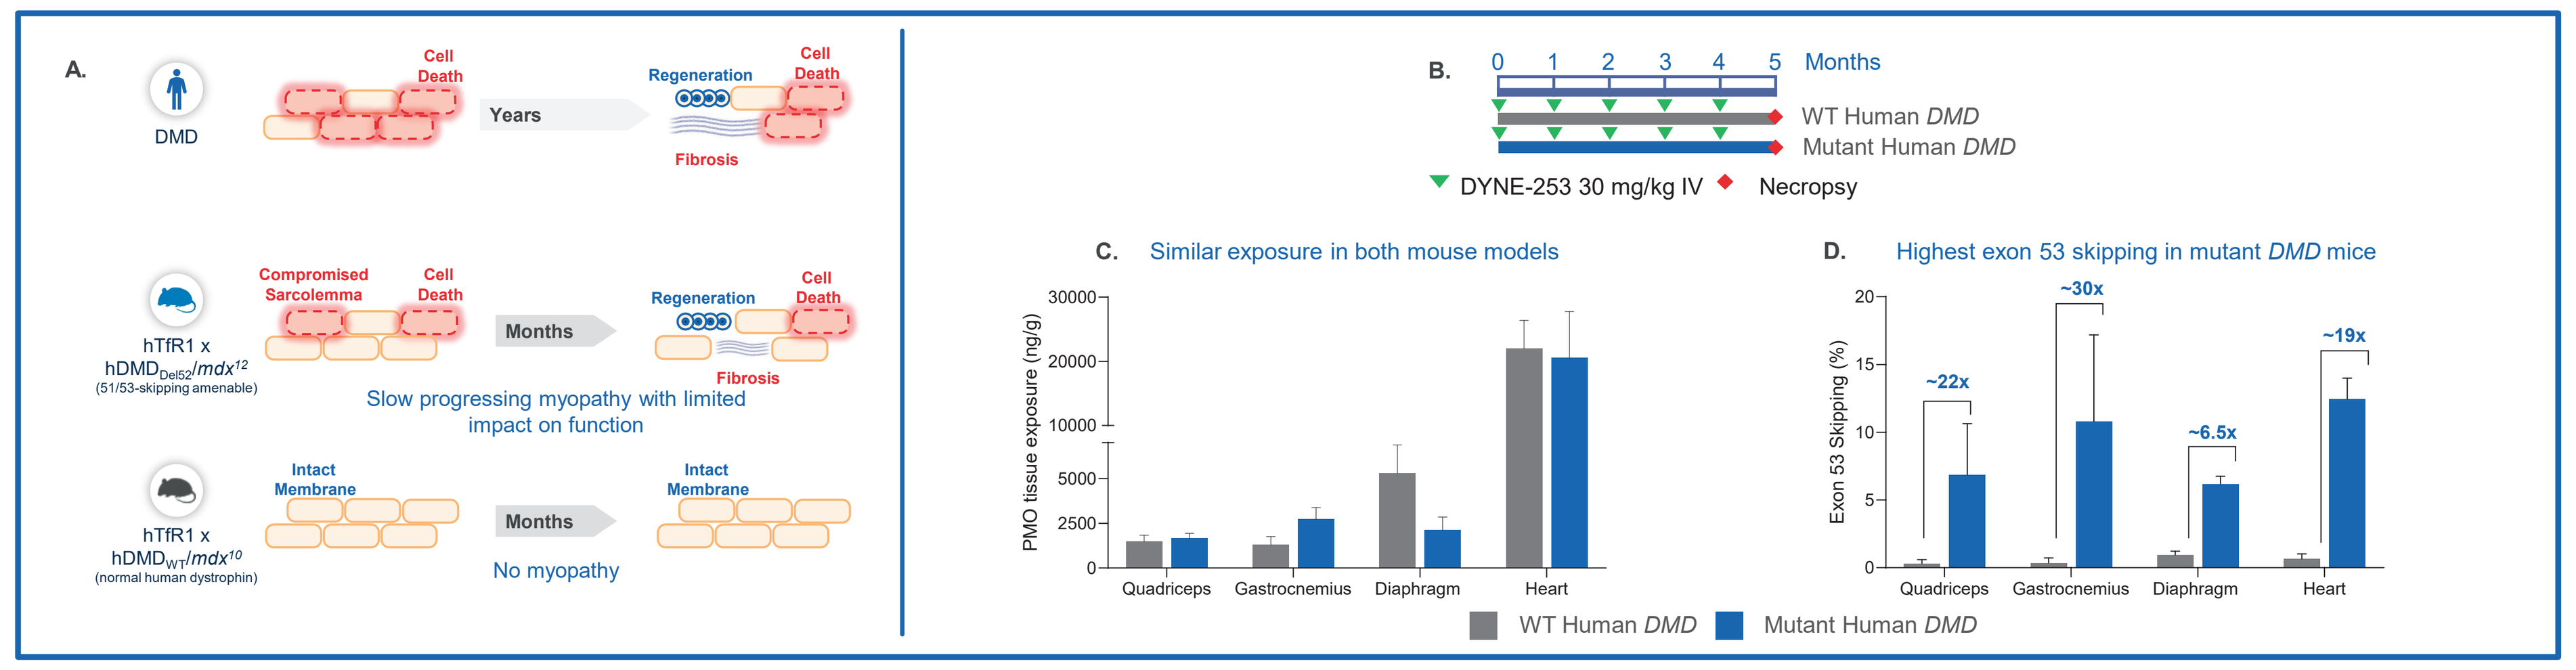
A.
DMD
Cell Death
Years
Regeneration
Fibrosis
Compromised Sarcolemma
Months
Slow progressing myopathy with limited impact on function
Intact Membrane
No myopathy
B. Months
WT Human DMD
Mutant Human DMD
DYNE-253 30 mg/kg IV
Necropsy
C. Similar exposure in both mouse models
PMO tissue exposure (ng/g)
D. Highest exon 53 skipping in mutant DMD mice
Exon 53 Skipping (%)
~22x
~30x
~6.5x
~19x
Quadriceps
Gastrocnemius
Diaphragm
Heart
WT Human DMD
Mutant Human DMD

The potency of an exon 53 skipping conjugate, DYNE-253, was assessed in humanized mouse models bearing a normal human *DMD* gene (hTfR1 x $hDMD_{WT}$/*mdx*) or a human *DMD* gene bearing a deletion of exon 52 (hTfR1 x $hDMD_{Del52}$/*mdx*). (A) Schematic representation of the phenotype and expected pathology of mouse models of interest. (B) Male mice of each genotype were administered DYNE-253 at a dose of 30 mg/kg PMO eq once monthly for 5 months, then necropsied for molecular assessments 1 month after the final dose. (C) Tissue exposure analysis showed substantial differences in the ability of DYNE-253 to deliver the exon skipping payload in either mouse model. (D) End-point PCR and capillary electrophoresis reveals substantially more exon 53 skipping in limb muscles, diaphragm, and heart of the hTfR1 x $hDMD_{Del52}$/*mdx* mouse model. Data are represented as mean ± SD, n=4 mice/group.

Figure 6. DYNE-253 Achieves Exon Skipping and Dystrophin Expression in Cardiac and Skeletal Muscle of hTfR1 x $hDMD_{Del52}$/*mdx* Mice

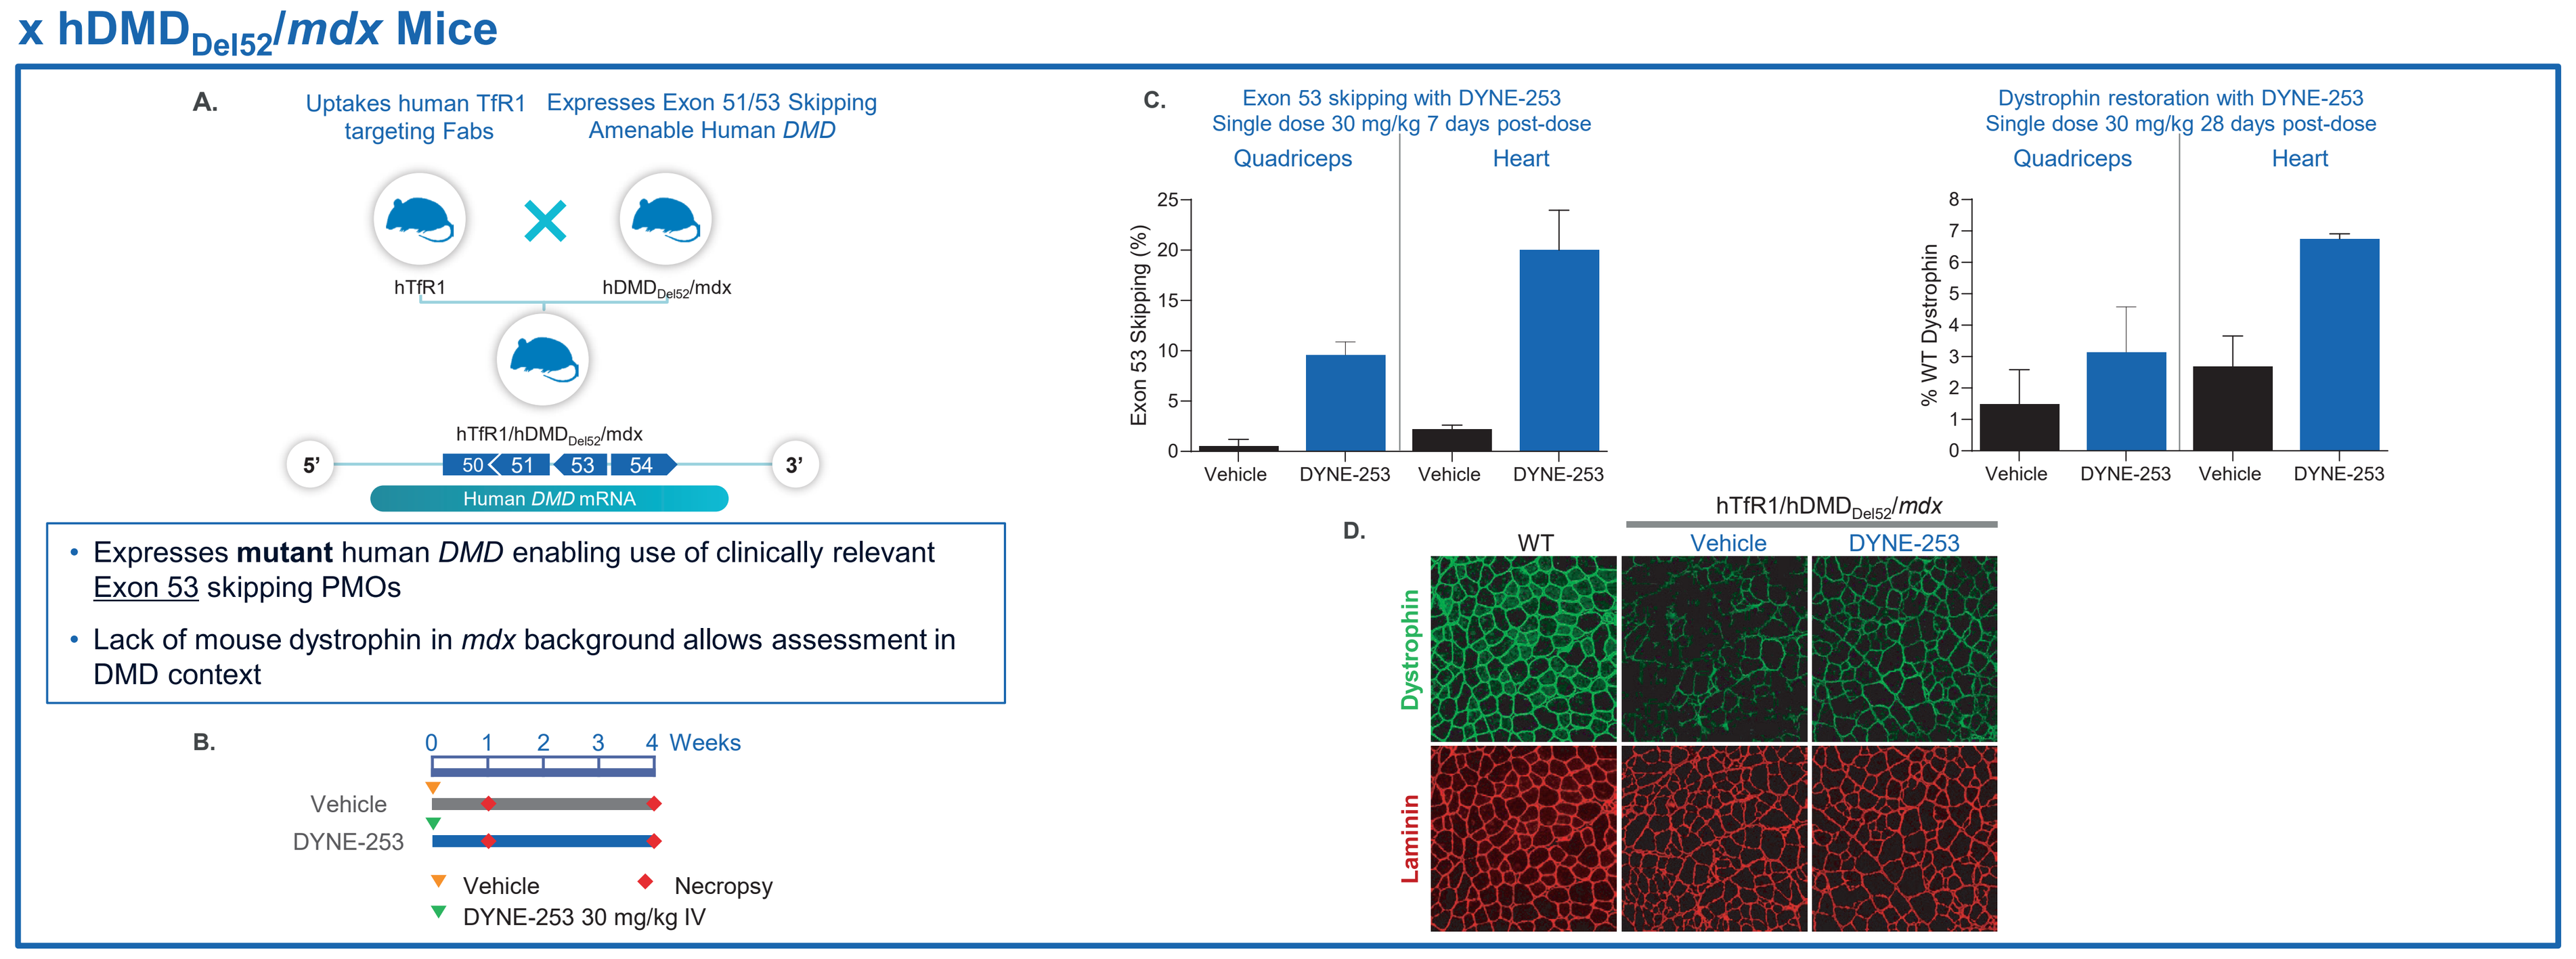
A. Uptakes human TfR1 targeting Fabs
Expresses Exon 51/53 Skipping Amenable Human DMD
hTfR1
hDMDDel52/mdx
hTfR1/hDMDDel52/mdx
Human DMD mRNA
Expresses mutant human DMD enabling use of clinically relevant Exon 53 skipping PMOs
Lack of mouse dystrophin in mdx background allows assessment in DMD context
B. Weeks
Vehicle
DYNE-253
Necropsy
DYNE-253 30 mg/kg IV
C. Exon 53 skipping with DYNE-253 Single dose 30 mg/kg 7 days post-dose
Dystrophin restoration with DYNE-253 Single dose 30 mg/kg 28 days post-dose
Quadriceps
Heart
Exon 53 Skipping (%)
% WT Dystrophin
D.
WT
Vehicle
Dystrophin
Laminin

A single dose of FORCE-253 induced exon 53 skipping and subsequent dystrophin protein expression and localization to the sarcolemma. (A) Schematic representation of the generation of humanized mice expressing a human TfR1 protein and a human *DMD* gene bearing a deletion of exon 52 that is amenable to exon 53 skipping. (B) Male hTfR1 x $hDMD_{Del52}$/*mdx* were administered a single IV dose of vehicle or 30 mg/kg PMO eq DYNE-253 and were necropsied 7 days or 28 days post-dose. (C) Exon 53 skipping was detectable via end-point PCR and capillary electrophoresis by 7 days post-dose (left graph) and induction of dystrophin protein expression by 28 days post-dose via semi-quantitative western blot (right graph). Data are represented as mean ±SD, n=2 mice/group for controls and n=3 mice/group for DYNE-253 treatment. (D) Representative immunofluorescence staining for dystrophin (top) and laminin (bottom) show induction of dystrophin protein expression in the hTfR1 x $hDMD_{Del52}$/*mdx* 28 days post-dose.

CONCLUSIONS

- All development candidates leverage the same FORCE™ platform as zeleciment rostudirsen (z-rostudirsen, also known as DYNE-251), which is under FDA review for DMD amenable to exon 51 skipping
- Data support the ongoing development of four potential therapies for DMD amenable to skipping of exons 53, 45, 44 or 55, all of which are advancing into IND-enabling studies
 - Robust exon skipping observed in disease-relevant myotubes for each exon skipping conjugate
 - The FORCE platform delivers substantially more payload to tissues of interest compared to unconjugated payload
 - Induction of exon 53 skipping in the hTfR1 x $hDMD_{Del52}$/*mdx* mouse led to dystrophin protein restoration and localization to the sarcolemma

REFERENCES

1. Muntoni F, Torelli S, Ferlini A. Dystrophin and mutations: one gene, several proteins, multiple phenotypes. Lancet Neurol. 2003 Dec;2(12):731-40.
2. Birnkrant DJ, Panitch HB, Benditt JO, Boitano LJ, Carter ER, Cwik VA, et al. American College of Chest Physicians consensus statement on the respiratory and related management of patients with Duchenne muscular dystrophy undergoing anesthesia or sedation. Chest. 2007 Dec;132(6):1977-86.
3. Cheeran D, Khan S, Khera R, Bhatt A, Garg S, Grodin JL, et al. Predictors of death in adults with Duchenne muscular dystrophy-associated cardiomyopathy. J Am Heart Assoc. 2017 Oct;6(10):e006340.
4. Naidoo M, Anthony K. Dystrophin Dp71 and neuropathophysiology of Duchenne muscular dystrophy. 2020 Mar;57(3):1748-1767.
5. Moulton HM, Moulton JD. Morpholinos and their peptide conjugates: therapeutic promise and challenge for Duchenne muscular dystrophy. Biochim Biophys Acta. 2010 Dec;1798(12):2296-303.
6. Saoudi, A., Fergus, C., Gileadi, T., Montanaro, F., Morgan, J.E., Kelly, V.P., Tensorer, T., Garcia, L., Vaillend, C., Muntoni, F. et al. (2023) Investigating the Impact of Delivery Routes for Exon Skipping Therapies in the CNS of DMD Mouse Models. Cells, 12.
7. Weeden T, Picariello T, Quinn B, et al. FORCE platform overcomes barriers of oligonucleotide delivery to muscle and corrects myotonic dystrophy features in preclinical models. *Commun Med (Lond)*. 2025;5(1):22. Published 2025 Jan 18. doi:10.1038/s43856-025-00733-w.
8. Charleston JS, Schnell FJ, Dworzak J, Donoghue C, Lewis S, Chen L, et al. Eteplirsen treatment for Duchenne muscular dystrophy: Exon skipping and dystrophin production. Neurology. 2018 Jun;90(24):e2146-e2154.
9. Polonifi A, Politou M, Kalotychou V, Xiromeritis K, Tsironi M, Berdoukas V, et al. Iron metabolism gene expression in human skeletal muscle. Blood Cells Mol Dis. 2010 Oct;45(3):233-7.
10. Gammella E, Buratti P, Cairo G, Recalcati S. The transferrin receptor: the cellular iron gate. Metallomics. 2017 Oct;9(10):1367-75.
11. Mayle KM, Le AM, Kamei DT. The intracellular trafficking pathway of transferrin. Biochim Biophys Acta. 2012 Mar;1820(3):264-81.
12. Veltrop M, van Vliet L, Hulsker M, et al. A dystrophic Duchenne mouse model for testing human antisense oligonucleotides. *PLoS One*. 2018;13(2):e0193289. Published 2018 Feb 21. doi:10.1371/journal.pone.0193289.

DISCLOSURE INFORMATION

All authors are employees or advisors of Dyne Therapeutics Inc. and may hold Dyne stock and/or stock options.

ACKNOWLEDGEMENTS

We would like to acknowledge the efforts of Amanda Clark from Biomere and Stephanie Rinaldi of Dyne Therapeutics in supporting animal care & in-life portions of these studies.

Poster Presented at 2026 Annual Neuromuscular Study Group Scientific Meeting